Versikor500® supplementation in palliative care of companion dogs suffering from malignant evolutive solid tumors

Agata Rybicka[1], Claire Mazuy[1], Quentin Pascal[1], Franck Floch[2], Laurie Boissy[2], Melanie Seffert[3], Virginie Coste[1], Dominique Tierny[1,2]

[1]OCR (Oncovet Clinical Research): Parc Eurasanté Lille Métropole, 80 Rue du Dr Yersin, 59120 Loos, France
[2]Oncovet: Avenue Paul Langevin, 59650 Villeneuve d'Ascq, France
[3] SumLabVet; 345 Montée de Bellevue, 01600 Reyrieux,France

Introduction

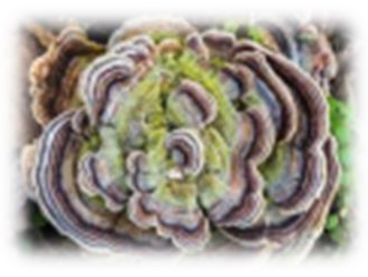

PSP: polysaccharopeptide extracted from Cov-1 strain of *Coriolus Versicolor* spawn.

In human

- **Used in traditional chinese medicine and in human clinical practice** in Asia as adjuvant in anti-cancer treatment
- Known as **immunomodulator** : ↑ IFNγ / IL-2 / TNFα (Lee, 2006; Sekhon, 2013; Hsieh, 2002)
- **Decreases the paraneoplasic effects and adverse reactions of chemotherapy** in human (Liu, 1993)

In dog

- **Delays abdominal metastasis progression** at 100 mg/kg/day
- **Prolongs median survival time** at 100 mg/kg/day : 199 days
- **Shows no adverse effects** due to treatment administration
- To date, only 1 published study on 15 dogs with resected splenic hemangiosarcoma (Brown, 2012)

Versikor500® is a food supplement enriched in PSP (>38% polysaccharides, >11,5% peptides) dedicated to dogs and cats

Aim of the study

Primary objective:
- Evaluate the **immunomodulatory effect of Versikor500®** on dogs suffering from solid malignant tumors

Secondary objectives:
- Evaluate the **impact of Versikor500® on dog quality of life** suffering from cancer and **on its owner quality of life**
- **Confirm the safety of Versikor500® administration** on dogs suffering from cancer

Exploratory objective:
- Evaluate the **overall survival of dog** receiving Versikor500®

Materials and methods

EXPERIMENTAL DESIGN

- **Multicentric non-comparative study** (12 investigation sites in France and Belgium)
- **22 dogs** suffering from **malignant evolutive solid tumors** (cytologically or histologically confirmed)
- Owners that refused surgery of the primary tumor or **any adjuvant treatment of residual disease (radiotherapy or chemotherapy)**
- Dogs not treated with immunosuppressive or analgesics treatments*

*** During the study, if required by the clinical status of a dog, maximum 5 days of corticosteroids or analgesic treatment (NSAIDs or Tramadol) was allowed**

STUDY DESIGN

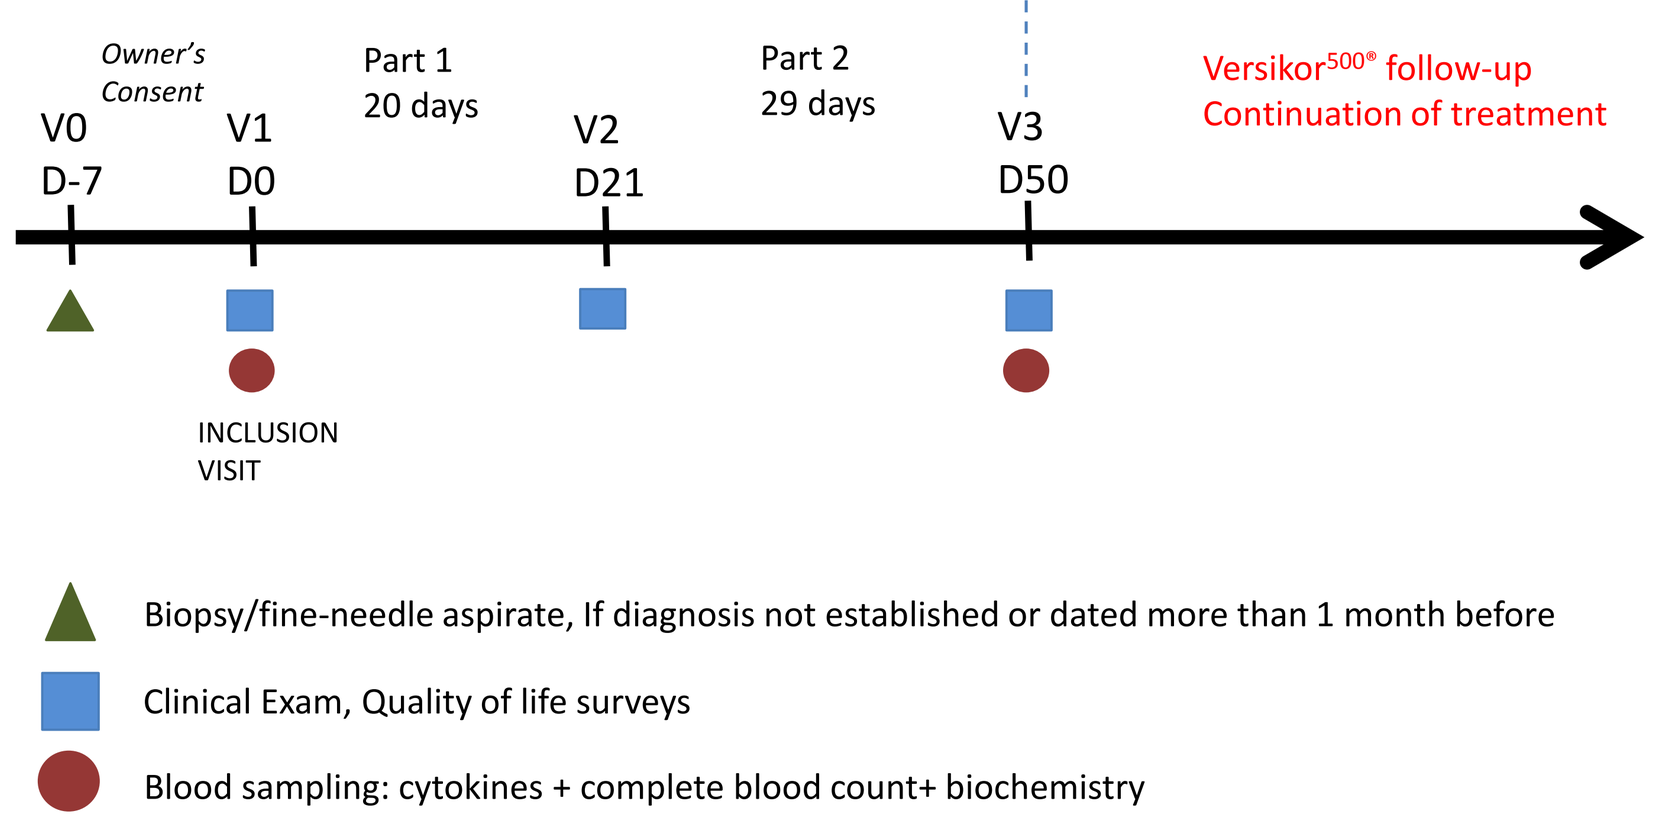
Owner's Consent
V0
D-7
V1
D0
Part 1
20 days
V2
D21
Part 2
29 days
V3
D50
Versikor500® follow-up
Continuation of treatment
INCLUSION VISIT
Biopsy/fine-needle aspirate, If diagnosis not established or dated more than 1 month before
Clinical Exam, Quality of life surveys
Blood sampling: cytokines + complete blood count+ biochemistry

STUDY TREATMENT

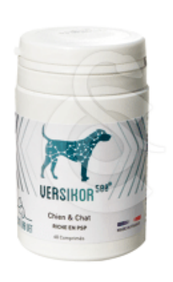

- 49 days of Versikor500® treatment at 100 mg/kg/day (500mg per tablet per 5kg body weight)
- Oral administration twice a day at home during the meal (morning and evening)

Complete Blood Count and Biochemistry parameters evaluation were performed by a central laboratory (Vebio, France). Cytokines assessment was performed in two batches using Luminex technology by the central laboratory.

Study Population

- 22 dogs included in the study
- **8 Males/ 14 Females**
 12 neutered / 10 not-neutered
- Mean age: 10,7 years old (6-16)
- Mean body weight (at V0): 23,5 kg (5,9-52)
- **16 dogs completed 50 days of study duration**
- Treatment compliance: Mean **98,3%**

Tumor types:
- **13 Sarcomas (59%):** various types, from very agressive (1 osteosarcoma, 3 hemangiosarcomas, 1 histiocytic sarcoma) to low grade soft tissue sarcomas
- **4 Mammary carcinomas (18,2%):** 2 tumors with grade 3 and 2 tumors with metastasis
- **3 Melanomas (13,7%):** 2 oral and 1 nail bed
- **2 Mastocytomas (9,1%):** grade 2

50% without surgery of the primary tumor
50% cases underwent surgery (22,7% had metastasis and 27,3% had residual disease)

Results

IMMUNOMODULATORY EFFECT

Analyses of immunomodulatory effect of Versikor500® were realized on the dogs which received the treatment for the entire study period and did not received concomitant treatment which could affect immune response.
Population of analysis : 10 dogs

PSP INCREASES PRO-INFLAMMATORY CYTOKINES SERUM CONCENTRATION

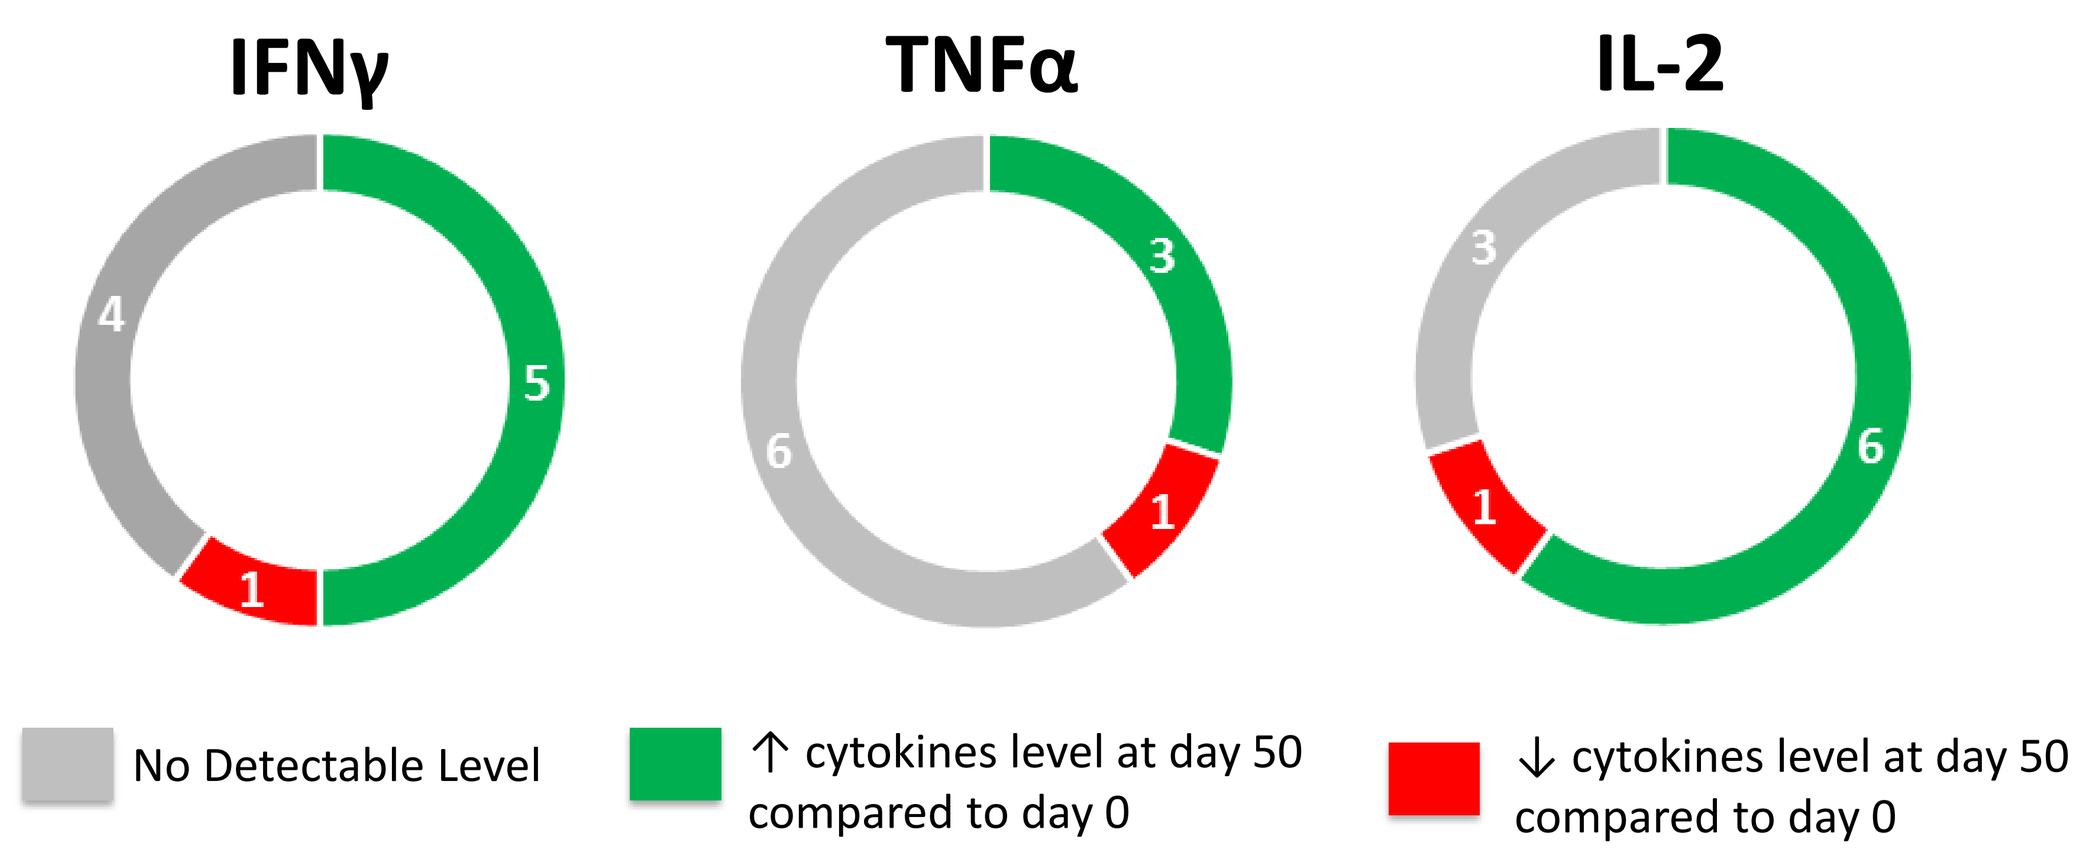
IFNγ
4
5
1
TNFα
3
6
1
IL-2
3
6
1
No Detectable Level
↑ cytokines level at day 50 compared to day 0
↓ cytokines level at day 50 compared to day 0

If we restrict the analysis to dogs with only detectable level of cytokines in serum, respectively **83%, 75% and 85% of dogs have shown increased level of IFNγ, TNFα and IL-2** in serum after 49 days of Versikor500® treatment.

MAINTENANCE OF LYMPHOCYTES BLOOD COUNT

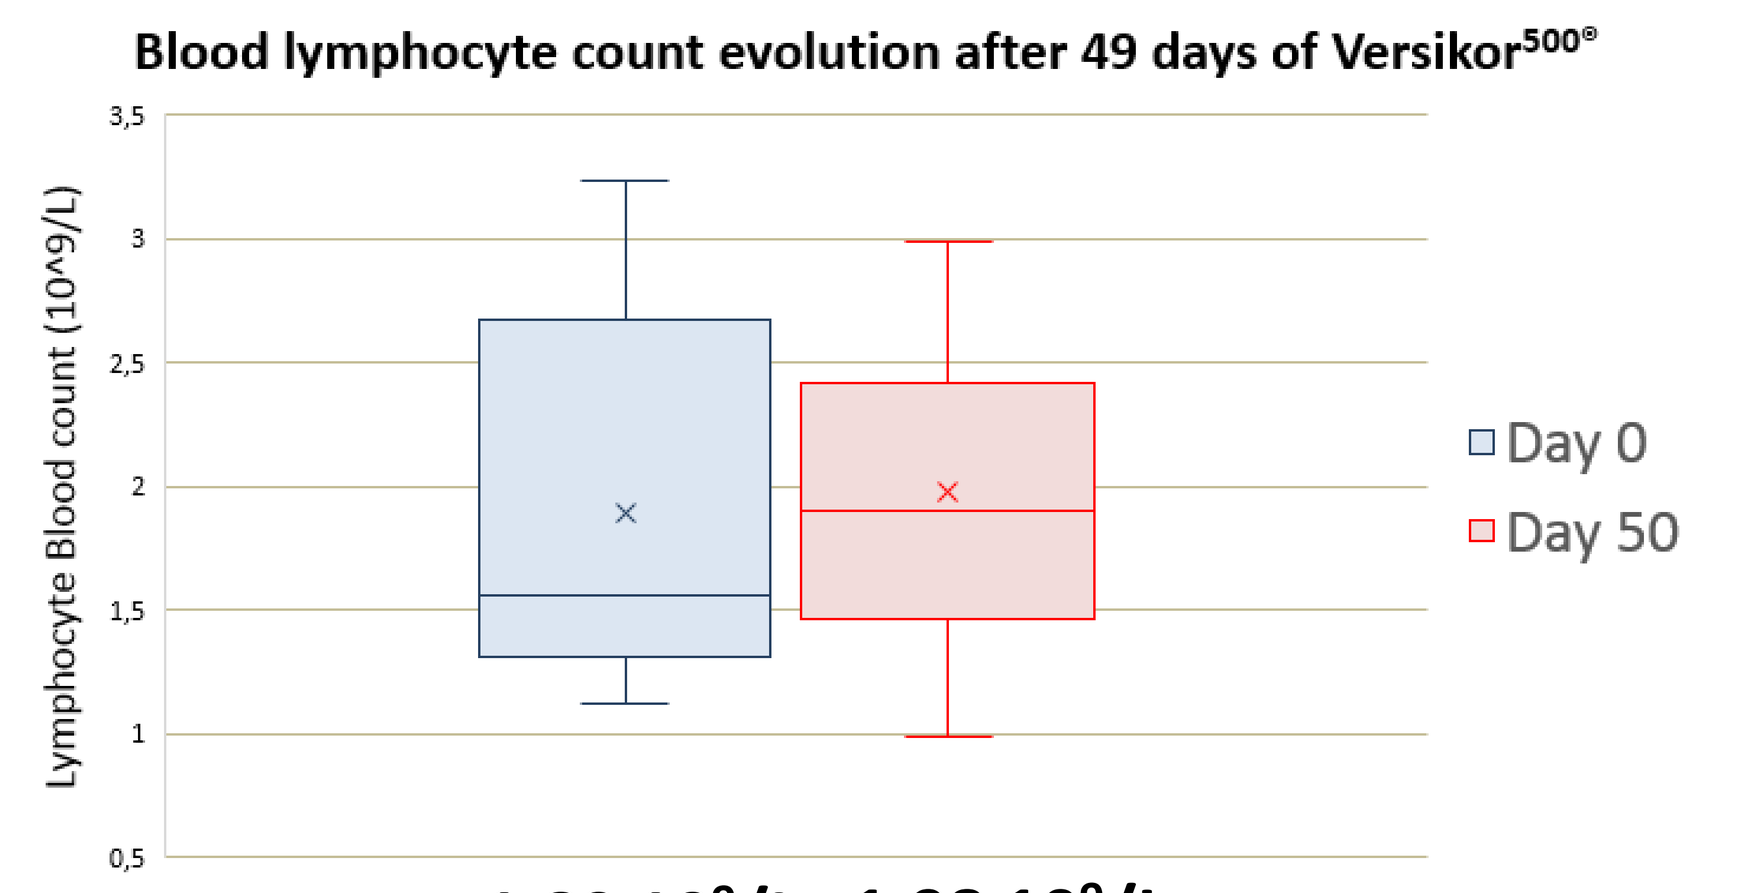
Blood lymphocyte count evolution after 49 days of Versikor500®
Lymphocyte Blood count (10^9/L)
Day 0
Day 50

	Day 0	Day 50
mean	1,89 10^9/L	1,98 10^9/L
median	1,50 10^9/L	1,91 10^9/L

Non-significant increase in blood lymphocyte count after 49 days of Versikor500®

DOG QUALITY OF LIFE

Analysis of dog quality of life was performed on dogs which received the treatment for the entire study period.
Decrease of quality of life score = improvement of dog quality of life
Population of analysis : 16 dogs

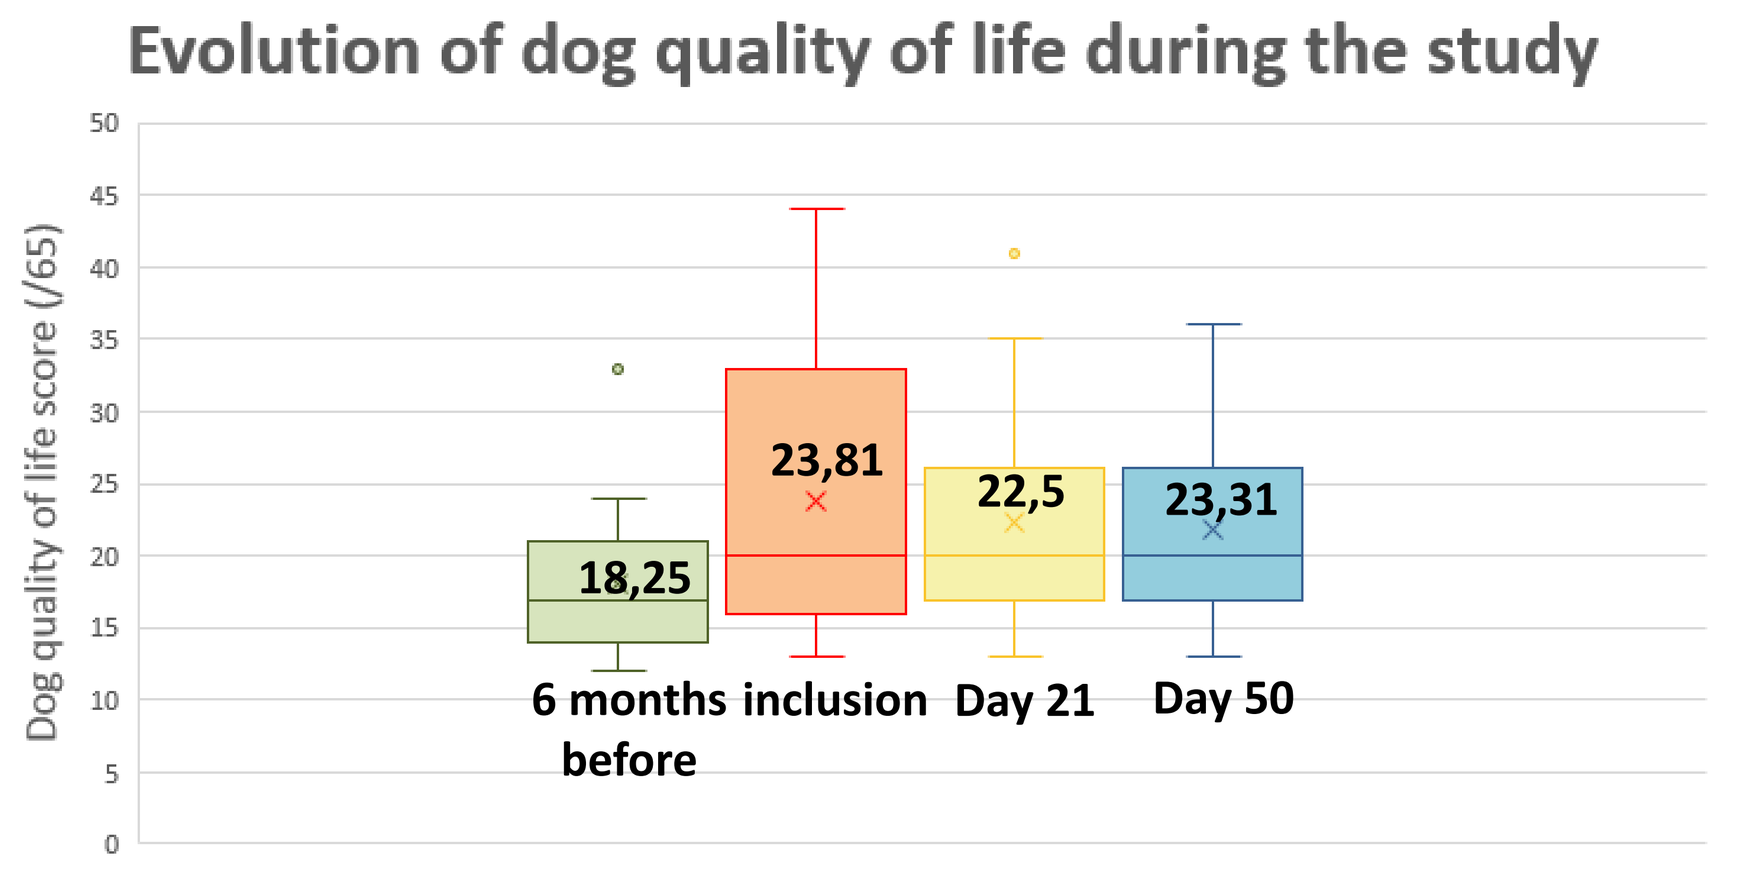
Evolution of dog quality of life during the study
Dog quality of life score (/65)
18,25
23,81
22,5
23,31
6 months before
inclusion
Day 21
Day 50

Maintenance of dog quality of life during the study duration

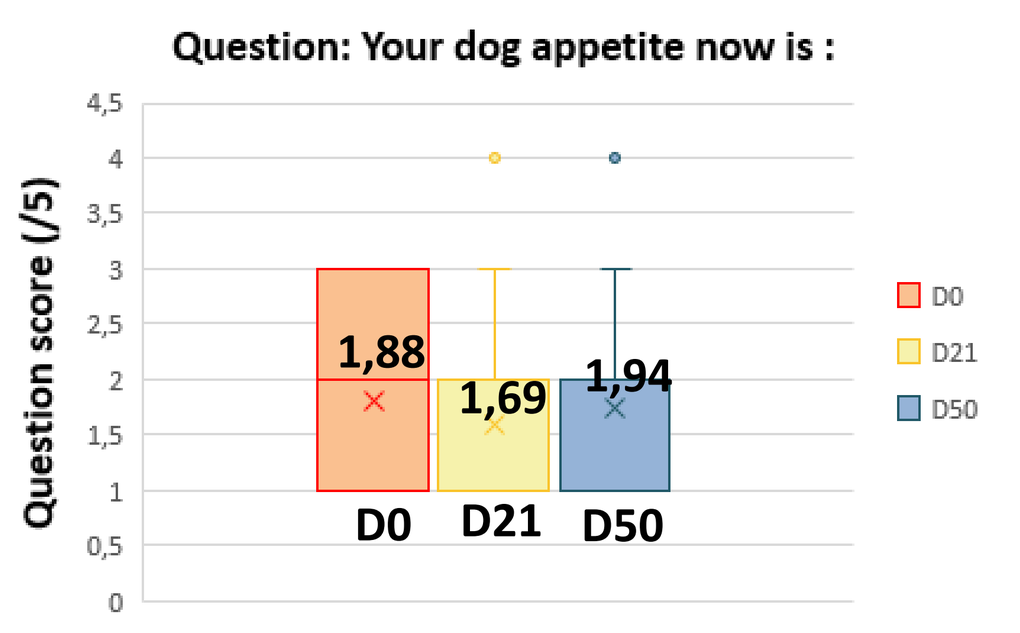
Question: Your dog appetite now is :
Question score (/5)
1,88
1,69
1,94
D0
D21
D50

Maintenance of dog appetite during the 50 days of treatment

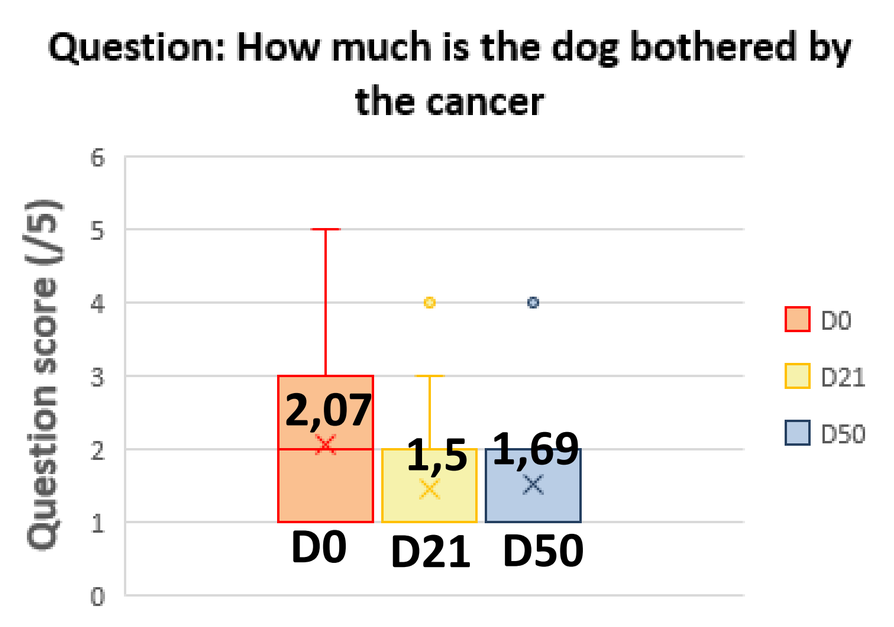
Question: How much is the dog bothered by the cancer
Question score (/5)
2,07
1,5
1,69
D0
D21
D50

Dogs are significantly less bothered by the cancer after 21 days of treatment compared to inclusion visit (Pv=0,02)

OWNER ANXIETY ABOUT DOG ILLNESS

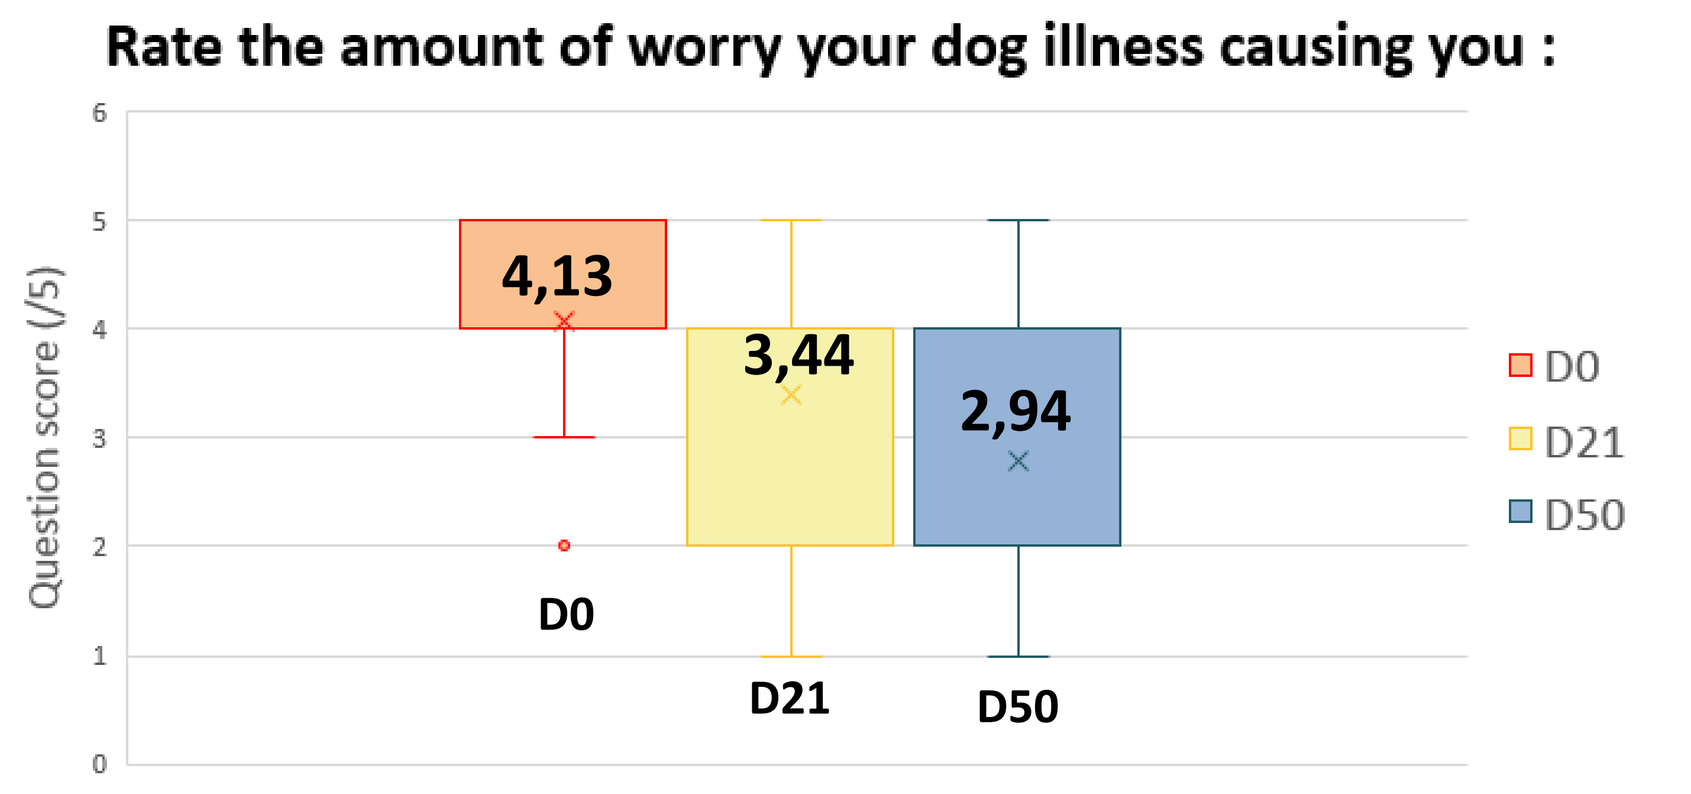
Rate the amount of worry your dog illness causing you :
Question score (/5)
4,13
3,44
2,94
D0
D21
D50

Significant **decrease of owner anxiety about dog illness** after 21 (Pv=0,02) and 50 days (Pv=0,005) of Versikor500® treatment.

SAFETY EVALUATION

- No difference in biochemical and haematological parameters
- No adverse events related or possibly related to the treatment
- Body weight / Rectal temperature stable during the study

5 SELECTED CASES WITH PROLONGED SURVIVAL

- 1 dog with **appendicular osteosarcoma - 124 days of survival without amputation**, euthanized due to severe pain (median survival time for OSA with amputation ≈133 days/ Spodnick GJ et al, 1992)
- 1 dog with **sub-cutaneous hemangiosarcoma (uncomplete exeresis) - 338 days of survival** on Versikor500®, natural dead (median survival time with or without cancer treatments is 172 days/ Shiu KB et al, 2011)
- **1 dog with mammary adenocarcinoma with hepatic metastasis and pancreatitis - 180 days of survival**, euthanized due to disease progression
- **1 dog with nail bed melanoma after surgery of the primary tumor with loco-regional lymph node metastasis - 308 days of survival**, still alive, in good health condition and still on Versikor500®
- **1 dog with multiple grade 2 mastocytoma 225 days of survival**, still alive and on Versikor500®

Conclusion

- The majority of dogs with assessable cytokines level, showed an **increase of IFNγ, IL-2 and TNFα serum concentration** after 49 days of Versikor500® treatment.
 ➔IL-2 is used in human medicine for its anti-tumoral effect, especially in kidney cancer with metastasis. The treatment is currently limited due to adverse effects of intravenous injection of this cytokine.
- Enrolled dogs **maintained their quality of life** during 50 days of study with **preservation of appetite**. Pet owners were less anxious by their dog illness.
- **No adverse events** were reported related to Versikor500® administration.
- Some cases presented prolonged survival

To sum up, **Versikor500® stimulates canine immune system and can be of added value in palliative care for various types of cancer.**
PSP is currently used as adjuvant to other anti-cancer treatments in human and demonstrates its ability to decrease their side effects and to maintain patients quality of life. Giving this effect, additional study with Versikor500 as adjuvant treatment would be of a major interest in dogs.

Acknowledgment

We would like to thank especially pet owners and investigators who actively participated in the case enrolment process for this study.
Versikor500® is still freely provided to study participants that are alive, if investigator and pet owner want to continue the treatment.

Contact

OCR : CEO Dr Dominique Tierny :
dtierny@oncovet-clinical-research.com
COO Mr Matthieu Dubruque :
mdubruque@oncovet-clinical-research.com
SUMLABVET : Dr Mélanie Seffert :
mseffert@sumlabvet.com

References

- Hsieh, T. C., Kunicki, J., Darzynkiewicz, Z., & Wu, J. M. (2002) *The Journal of Alternative & Complementary Medicine*
- Brown, D.C, Reetz, J. (2012) *Evid Based Complement Alternat Med*
- Lee, C. L., Yang, X., & Wan, J. M. F. (2006) *Enzyme and Microbial Technology*
- Liu, J. X., & Zhou, J. Y. (1993) *PSP International Symposium* Fudan University Press Hong-Kong
- Sekhon, B. K., Sze, D. M. Y., Chan, W. K., et al (2013) *Food Chemistry*
- Spodnick, G.J., Berg J., Rand W.M., et al (1992) *J Am Vet Med Ass*
- Shiu, K.B., Flory, A.B., Anderson, C.L., et al (2011) *J Am Vet Med Ass*